The new European regulation on Health Technology Assessment: what changes for Advanced Therapy Medicinal Products ?

Blanka Bartos, Joséphine Juët, Florence Taboulet, Aurélie Mahalatchimy

To cite this version:

Blanka Bartos, Joséphine Juët, Florence Taboulet, Aurélie Mahalatchimy. The new European regulation on Health Technology Assessment: what changes for Advanced Therapy Medicinal Products ?. European Society for Gene & Cell Therapy Congress 2022, Oct 2022, Edinburgh, United Kingdom. halshs-03901269

HAL Id: halshs-03901269
https://shs.hal.science/halshs-03901269

Submitted on 15 Dec 2022

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Distributed under a Creative Commons Attribution 4.0 International License

The new European regulation on Health Technology Assessment: what changes for Advanced Therapy Medicinal Products?

Blanka Bartos[1], **Joséphine Juët**[2], **Florence Taboulet**[3], **Aurélie Mahalatchimy**[4]

[1] PhD Student in law, CREDIMI, Univ. of Burgundy, Dijon, 27877, France, [2] Student in Pharmaceutical Sciences, Univ. of Paul Sabatier- Toulouse 3, Toulouse, 31062, France., [3] Professor in Pharmaceutical Law and Health Economics, CERPOP (Center for Epidemiology and Research in POPulation health), UMR 1295 Inserm, Univ. of Paul Sabatier- Toulouse 3, Toulouse, 31062, France., [4] Permanent Researcher in law at the French National Centre for Scientific Research (CNRS), Faculty of Law, UMR 7318 International, Comparative and European law (DICE) CERIC, CNRS, Aix Marseille Univ., Toulon Univ. Pau & Pays de l'Adour Univ., Aix-en-Provence, 13628, France

Acknowledgement: This work has been supported by ANR-funded I-BioLex project (ANR-20-CE26-0007-01, coord. A. Mahalatchimy).

Health Technology Assessment (HTA)?

"A **multidisciplinary process** that summarises information about the medical, patient and social aspects and the economic and ethical issues related to the use of a health technology in a systematic, transparent, unbiased and robust manner."

Article 2.5) EU Regulation 2021/2282

Evidence-based process enabling relative effectiveness assessment of new/ existing technologies

Health technology?

"A **medicinal product**, a medical device or medical and surgical procedures as well as measures for disease prevention, diagnosis or treatment used in healthcare."

Article 3.l) Directive 2011/24/EU

ATMPs are included

PAST

- Independent assessments from national HTA bodies
- EUnetHTA (2006-2021): joint action, voluntary cooperation

BUT

- Redundancy of the evaluation process in the EU
- Misallocation of resources
- Inequitable access to new technologies

PRESENT (2022)

Rolling implementation plan: preparatory phase

- Setting up the Coordination Group and the Stakeholder Network
- Drafting implementing & delegated acts, and guidance documents

- 2021-23: New EunetHTA (2021)
- 2023-25: Member States remain responsible for drawing conclusions on added value for their health system and taking decisions on pricing & reimbursement. They can give additional multinational or simultaneous national scientific advice on a voluntary basis.

FUTURE (from 2025)

Application of Joint Clinical Assessments:

- **2025: ATMPs, cancer drugs**
- 2028: orphan drugs
- 2030: all medicines covered by Regulation scope

New HTA process:

- New **Coordination Group** managing HTA process
- **Secretariat** provided by the European Commission
- **National bodies** providing expert views on HTA
- Exchange of information with **stakeholder network**

- Increased quality of joint work
- Better allocation of resources: reduced expenses for national bodies and the EU
- Improved accessibility to ATMPs in the EU

Clinical and non-clinical assessments

Joint Clinical Assessments → To be considered for any HTA at Member States level

Description	Goal
Collection and analysis of clinical evidence about a health technology to determine its therapeutic value compared to other technologies already available or not.	Quality, common source for States to determine pricing and reimbursement of health technology: **ATMPs+++**

Voluntary Cooperation to exchange information, e.g., on non-clinical assessments

Description	Goal
Allowing States to exchange information on non-clinical areas, for examples on organizational, ethical, legal, and economic matters, or on health technologies other than medicines and medical devices	Going beyond the clinical scope: **ATMPs+++**

Complementary tools

Support to industry

Joint Scientific Consultations → No legal effects on Member States

Description	Goal
Early exchanges between industry and the Coordination Group.	Advising on clinical studies designs to generate appropriate evidence: **ATMPs+++**

Support to national governments

Identification of Emerging Technologies (ATMPs) → Coordination Group's Initiative

Description	Goal
Reports on new technologies with potentially significant impact on patients, public health and healthcare systems.	Anticipating issues to improve patient access: **ATMPs+++**

Remaining challenges:

- Building EU processes from national HTA experiences and feedback from experts
- Training more external expert / organisations on HTA in general, and on EU procedure(s)
- Organising HTA registries, raising awareness, collecting experiences
- Identifying / involving clinicians or patients (organisations' membership list, social networks, European Reference Networks, clinics in Pubmed...)

EUROPEAN SOCIETY OF GENE & CELL THERAPY

Edinburgh 2022 29th Congress in collaboration with British Society for Gene & Cell Therapy 11 to 14 October